rhGM-CSF (molgramostim) is being developed for the treatment of ARDS and Pulmonary Alveolar Proteinosis - This is Phase I

SAFETY, TOLERABILITY, PHARMACOKINETICS AND PHARMACODYNAMICS OF INHALED MOLGRAMOSTIM, A RECOMBINANT HUMAN GRANULOCYTE MACROPHAGE-COLONY STIMULATING FACTOR (RHGM-CSF), IN AN ASCENDING SINGLE AND MULTIPLE-DOSE TRIAL IN HEALTHY SUBJECTS

Tarnow I, Nymark K, Vinge M, Nielsen KA, Ganslandt C

Serendex Pharmaceuticals A/S, Slotsmarken 17, 2.tv, DK-2970 Hørsholm, Denmark

INTRODUCTION

GM-CSF is a cytokine and a hematopoietic growth factor produced by a variety of cells. In the lungs GM-CSF regulates surfactant homeostasis and lung host defense through innate immune functions. Alveolar macrophages from GM-CSF-/- mice have reduced capacity for surfactant catabolism, cell adhesion, phagocytosis and bacterial killing. Disruption of surfactant homeostasis by autoantibodies to GM-CSF cause autoimmune pulmonary alveolar proteinosis, an ultra-rare disease in which clinical effect of exogenous rhGM-CSF has been documented in several case reports and uncontrolled clinical trials. GM-CSF is highly protective in non-clinical models of pneumonia-associated lung injury, where GM-CSF protects the host in both early phase of acute lung infection and during regeneration phase. Molgramostim is an un-glycosylated rhGM-CSF produced in a strain of Escherichia coli bearing a genetically engineered plasmid which contains a human GM-CSF gene. A nebuliser solution containing molgramostim has been formulated and developed specifically for inhalation treatment of respiratory conditions

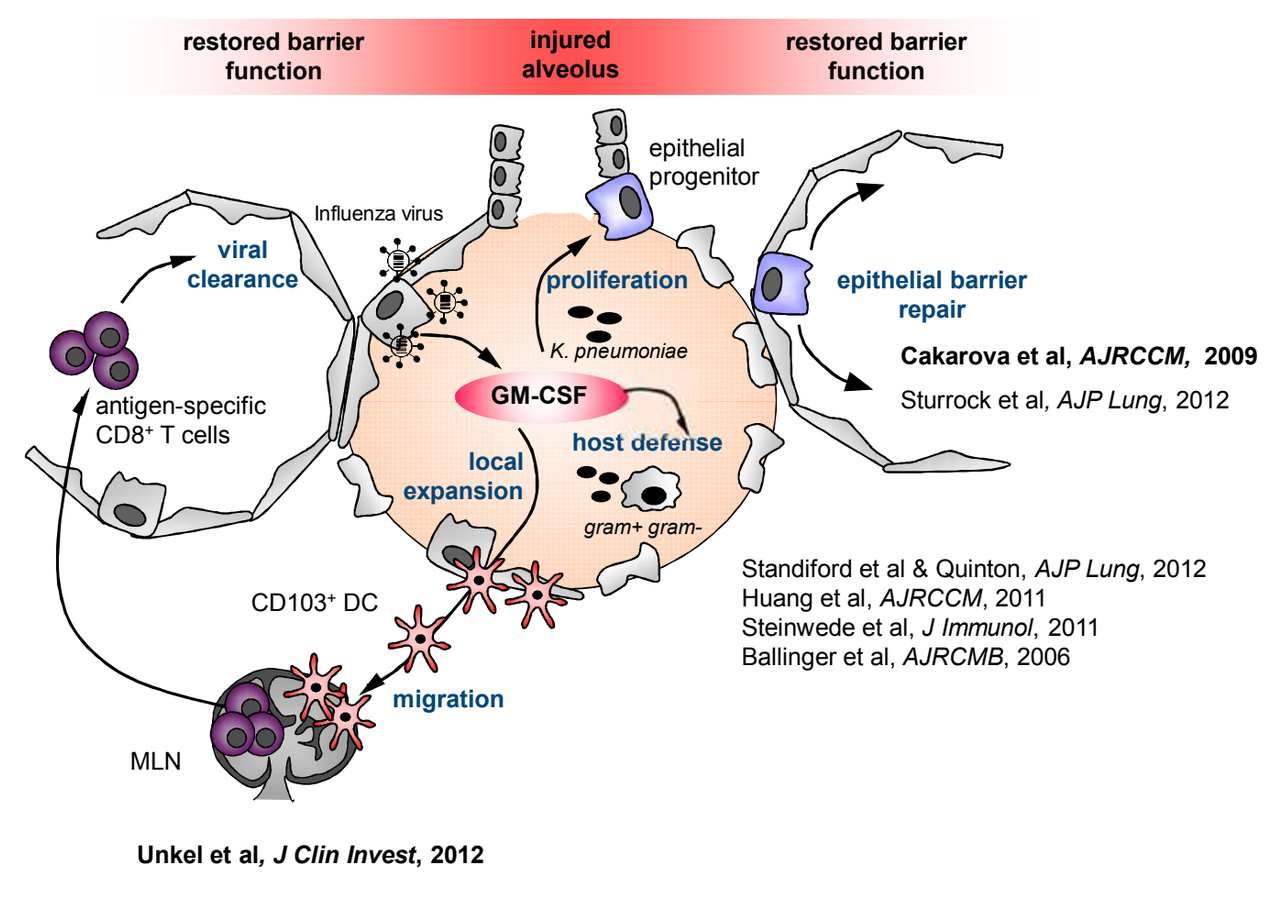
restored barrier function
injured alveolus
restored barrier function
epithelial progenitor
Influenza virus
viral clearance
proliferation
K. pneumoniae
epithelial barrier repair
Cakarova et al, AJRCCM, 2009
Sturrock et al, AJP Lung, 2012
antigen-specific CD8+ T cells
GM-CSF
host defense
local expansion
gram+ gram-
CD103+ DC
Standiford et al & Quinton, AJP Lung, 2012
Huang et al, AJRCCM, 2011
Steinwede et al, J Immunol, 2011
Ballinger et al, AJRCMB, 2006
migration
MLN
Unkel et al, J Clin Invest, 2012

Picture courtesy of Professor S. Herold, University of Giessen-Marburg Lung Center, Germany

OBJECTIVES

1 Primary objective:
To assess the safety and tolerability of single and multiple ascending inhaled doses of molgramostim when administered to healthy adult subjects

2 Secondary and exploratory objectives:
To assess the pharmacokinetics (PK) of inhaled molgramostim when administered to healthy adult subjects

To assess the effect on white blood cell (WBC) differentials

CONCLUSIONS

Inhaled molgramostim is well-tolerated in healthy subjects, is absorbed systemically at picogram levels, and shows a clear effect on White Blood Cell (WBC) differentials mostly within normal reference ranges. Inhaled molgramostim may be a promising new treatment for respiratory conditions with a high unmet medical need

FUTURE WORK

Clinical trials of inhaled molgramostim have been initiated in 2016

ARDS (Phase II) to be conducted at two centers in Germany (NCT02595060)

Autoimmune pulmonary alveolar proteinosis (Phase II/III pivotal trial) to be conducted at 16 centres in Europe, Israel, Russia and Japan (NCT02702180)

REFERENCES

Trapnell and Whitsett. Annu Rev Physiol 2002. 64: 775-802, Trapnell et al. Curr Opin Immunol 2009. 21: 514-521,
Unkel et al. J Clin Invest 2012. 122: 3652-3664, Cakarova et al. Am J Respir Crit Care Med 2009. 180: 521-532
Herold et al. Am J Respir Crit Care Med 2014. 189: 609-611

SERENDEX
Pharmaceuticals

FORMULATION AND DELIVERY TO THE LUNGS

Molgramostim nebuliser solution is a liquid formulation containing molgramostim 250 µg/mL and is delivered using eFlow® nebuliser system (PARI Pharma GmbH, Germany)

The eFlow® handset is a single patient use, reusable nebuliser, which has been optimised for nebulisation of molgramostim nebuliser solution:

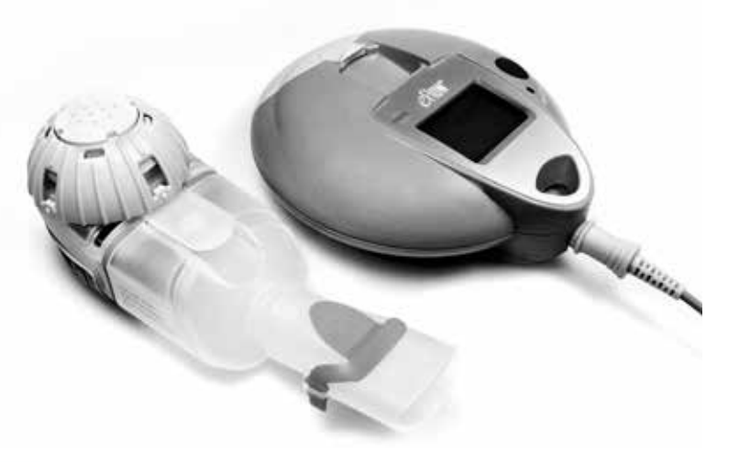

Parameter	Results
Results from laser diffraction	
Particle size distribution	Mass Median Diameter: 3.5 µm (SD 0.37) Geometric Standard Deviation: 1.6 (SD 0.1) Respirable fraction < 5 µm: 76.9% (SD 8.03)
Results from breath simulation experiments	
Delivered dose	60.4%
Respirable dose < 5 µm	46.3%

METHODS

The trial adhered to the principles of the Declaration of Helsinki and Good Clinical Practice. The trial was approved by The Medicines and Healthcare Products Regulatory Agency (UK) and Office for Research Ethics Committees, Northern Ireland, UK, and conducted at a specialized Phase I clinic in Belfast, UK (Celerion Inc.)

Study design
- Randomised, double-blind, placebo-controlled, single and multiple-dose trial
- Single ascending dose part (SAD) – 150 µg, 300 µg og 600 µg molgramostim (n=4 per dose level) or placebo (n=6) daily
- Multiple ascending dose part (MAD) - 300 µg og 600 µg molgramostim (n=9 per dose level) or placebo (n=6) daily for 6 days
- Follow-up visits 8 and 15 days after last dose
- Dose escalation only after review of safety and tolerability findings of preceding dose
- All dose levels in the SAD part included a sentinel group of 2 subjects
- Dose selection based on previous clinical experience with inhaled rhGM-CSF

Subjects
- All provided written informed consent
- 2 females and 40 males aged 18 – 53 years, Body mass index (BMI) ≥ 20.5 and ≤ 31.7 kg/m² at screening
- Life-long non-smoker, not using nicotine-containing products
- Healthy assessed by medical history, physical examination, spirometry, ECG, vital signs, and clinical laboratory test

RESULTS – SAFETY

No treatment-related effects on
- Vital signs including pulse oximetry
- Spirometry
- ECG (including Holter monitoring)
- Serum chemistry, and coagulation

No development of anti-molgramostim antibodies

Adverse event (AE) rates similar across groups, although higher number of events in 600 µg dose group in MAD part

Overview of reported Adverse Events (AEs) by treatment, only AEs reported in 2 or more subjects or AEs rated as related to treatment by the Principal Investigator (PI) are listed here

	Part 1 (SAD)				Part 2 (MAD)		
Treatment **Number of subjects**	**150 µg** **N=4**	**300 µg** **N=4**	**600 µg** **N=4**	**Placebo** **N=6**	**300 µg** **N=9**	**600 µg** **N=9**	**Placebo** **N=6**
No. (%) of Subjects Reporting AEs	2 (50%)	4 (100%)	1 (25%)	4 (67%)	7 (78%)	8 (89%)	6 (100%)
No. of AE Episodes	4	13	3	7	64	103	59
AE	Number of subjects reporting AE						
Cough*	1	4	1	2	7	8	6
Productive cough*	0	0	0	1	1	2	2
Throat irritation*	0	0	0	0	0	0	2
Dizziness#	0	0	0	0	2	2	2
Headache*	0	0	0	0	5	2	1
Eosinophilia*	0	0	0	0	0	1	0
Mild WBC increase*	0	0	0	0	0	1	0
Diarrhoea*	1	0	0	1	0	0	0
Tachycardia*	0	0	0	0	0	1	0

* Assessed as possibly, probably or likely related to blinded treatment by PI
Assessed as possibly related in one subject treated with 600 µg molgramostim, the AEs in the remaining 3 subjects were unrelated

RESULTS – PK

Molgramostim was absorbed from the lungs into the circulation resulting in serum GM-CSF concentration at picogram levels. GM-CSF in serum measured by electrochemiluminescence GLP-validated assay (Mesoscale), optimized to detect molgramostim in human serum

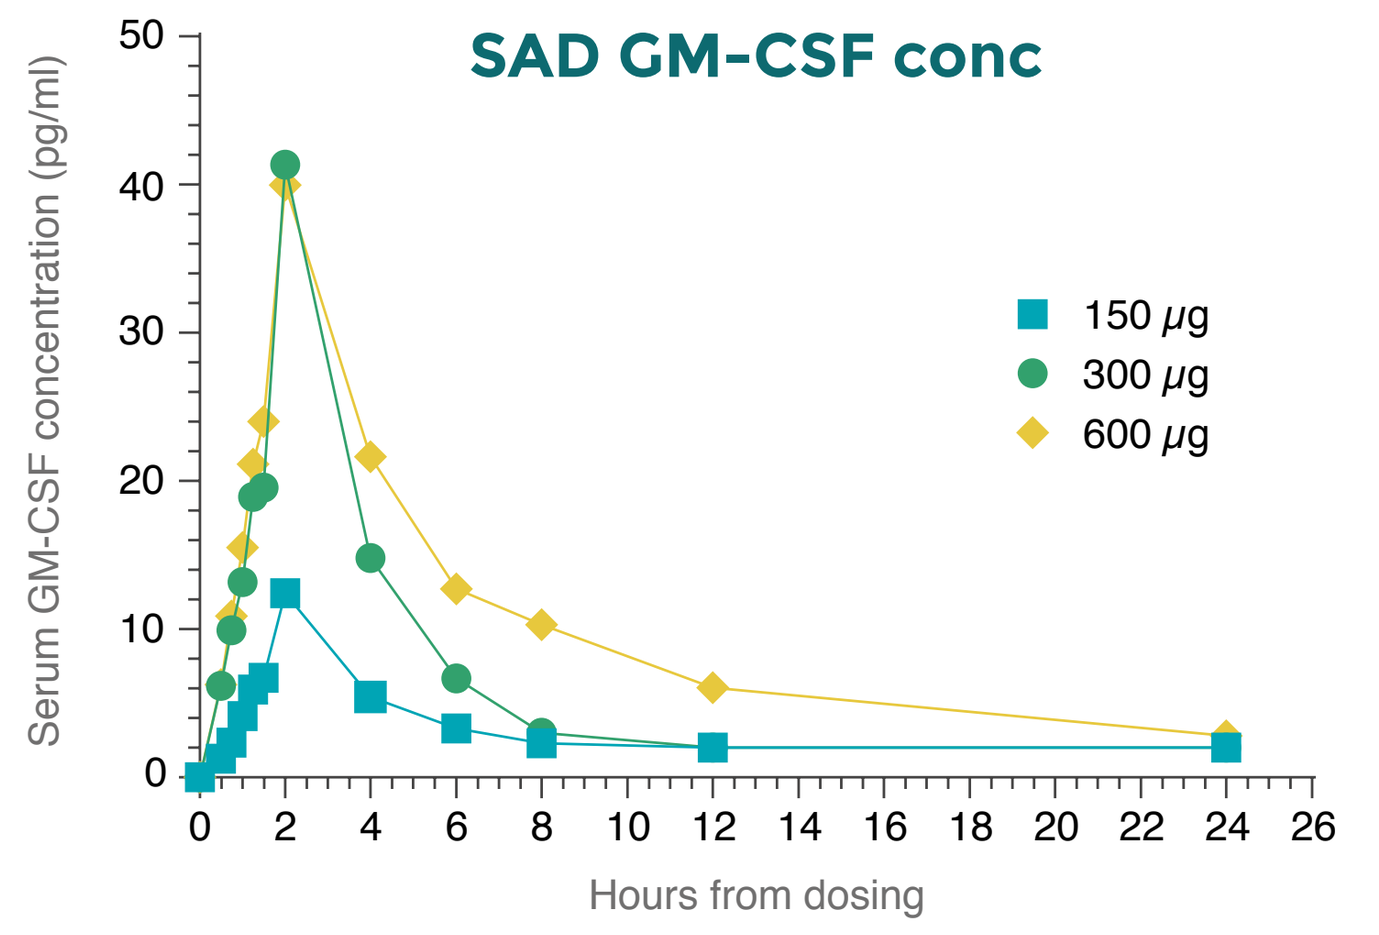
SAD GM-CSF conc
Serum GM-CSF concentration (pg/ml)
Hours from dosing
150 µg
300 µg
600 µg

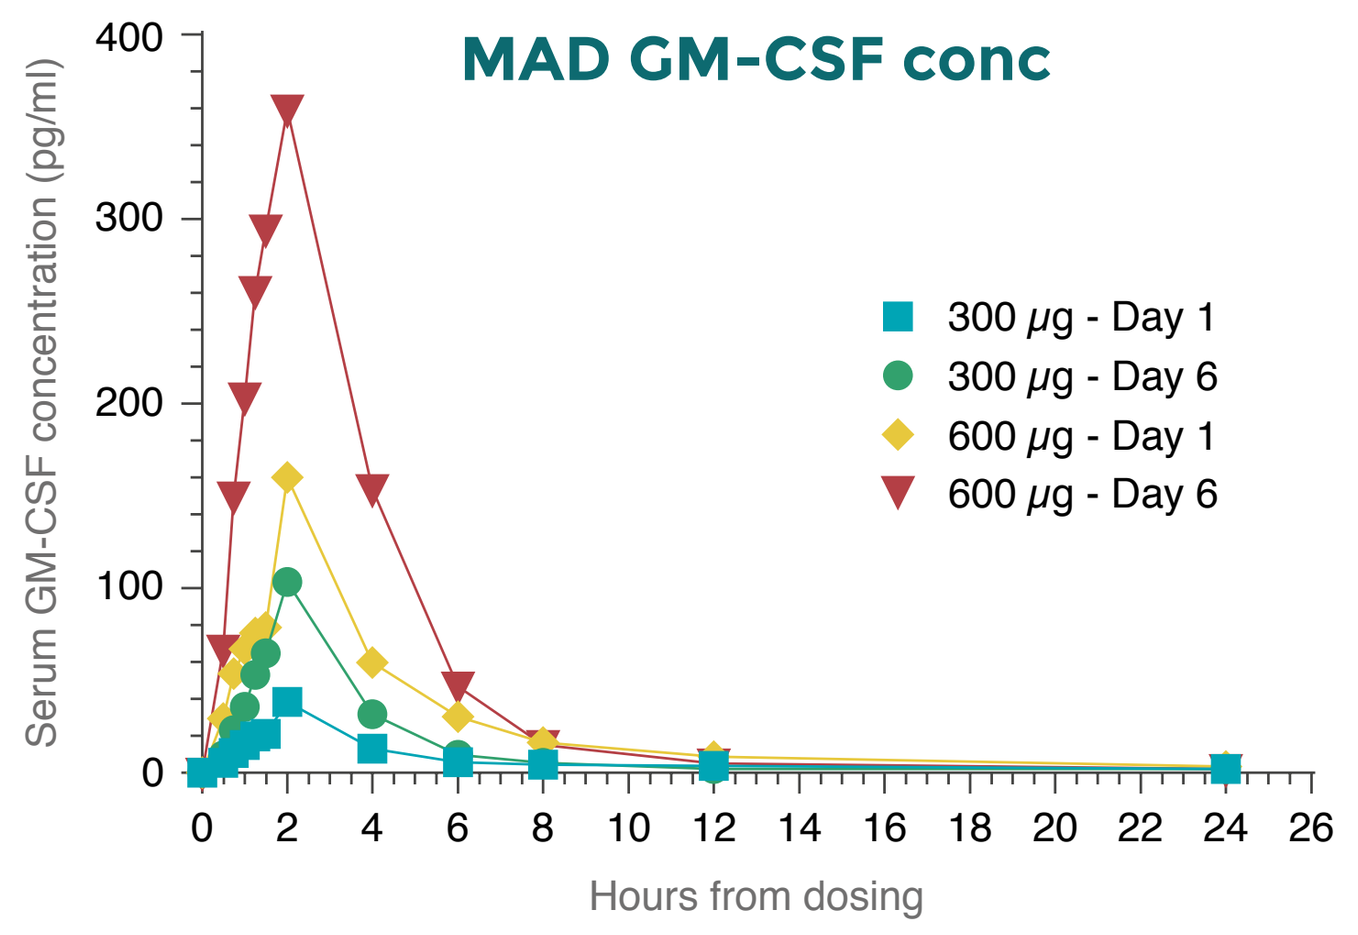
MAD GM-CSF conc
Serum GM-CSF concentration (pg/ml)
Hours from dosing
300 µg - Day 1
300 µg - Day 6
600 µg - Day 1
600 µg - Day 6

	Part 1 (SAD)			Part 2 (MAD)			
Parameter	**150 µg**	**300 µg**	**600 µg**	**300 µg Day 1**	**600 µg Day 1**	**300 µg Day 6**	**600 µg Day 6**
C_{max} (pg/mL)[a]	9.07 (116)	40.7 (57.4)	34.1 (73.6)	32.1 (73.4)	90.1 (63.4)	96.4 (139)	251 (109)
T_{max} (hr)[b]	2.01 (2.00, 2.01)	2.00 (1.25, 2.00)	2.00 (2.00, 2.00)	2.00 (2.00, 2.01)	2.01 (2.00, 2.04)	2.00 (2.00, 2.02)	2.02 (1.96, 2.10)
$AUC_{0\text{-}last}$ (pg*hr/mL)[a]	12.5 (662)	85.9 (100)	138 (122)	97.1 (72.5)	248 (53.5)	350 (125)	802 (119)
$T_{1/2}$ (hr)[c]	1.82	1.73 ± 0.0398	5.91 ± 0.854	3.95 ± 1.97	-	3.68 ± 0.784	-

a: Geometric mean (CV%)
b: Median (min, max)
c: Arithmetic mean ± SDev

RESULTS – WBC

Inhaled molgramostim resulted in dose-dependent, temporary increases from baseline in WBC, neutrophils, monocytes, and eosinophils

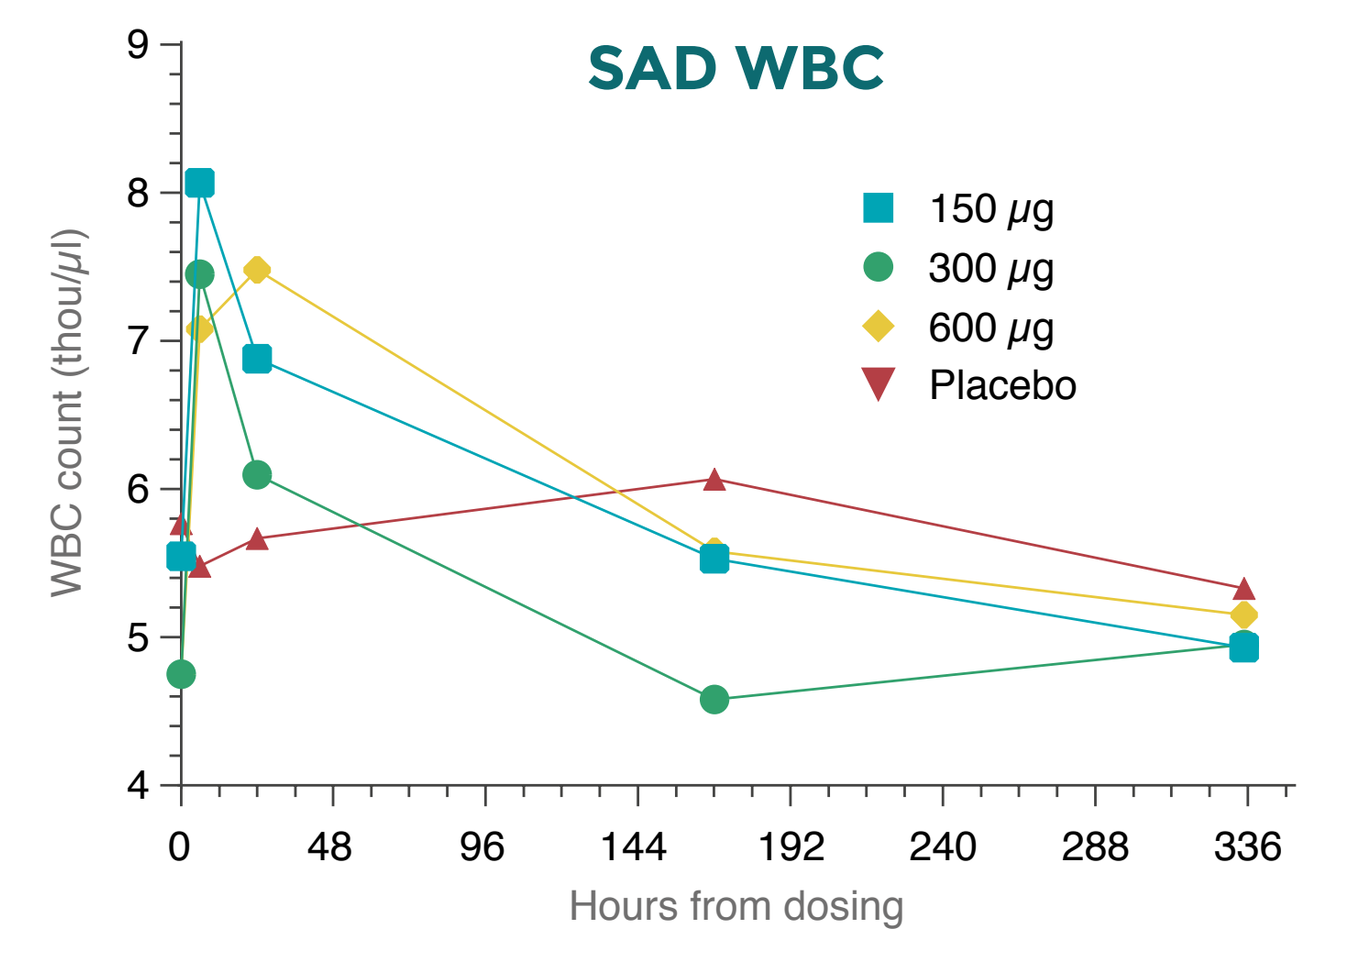
SAD WBC
WBC count (thou/µl)
Hours from dosing
150 µg
300 µg
600 µg
Placebo

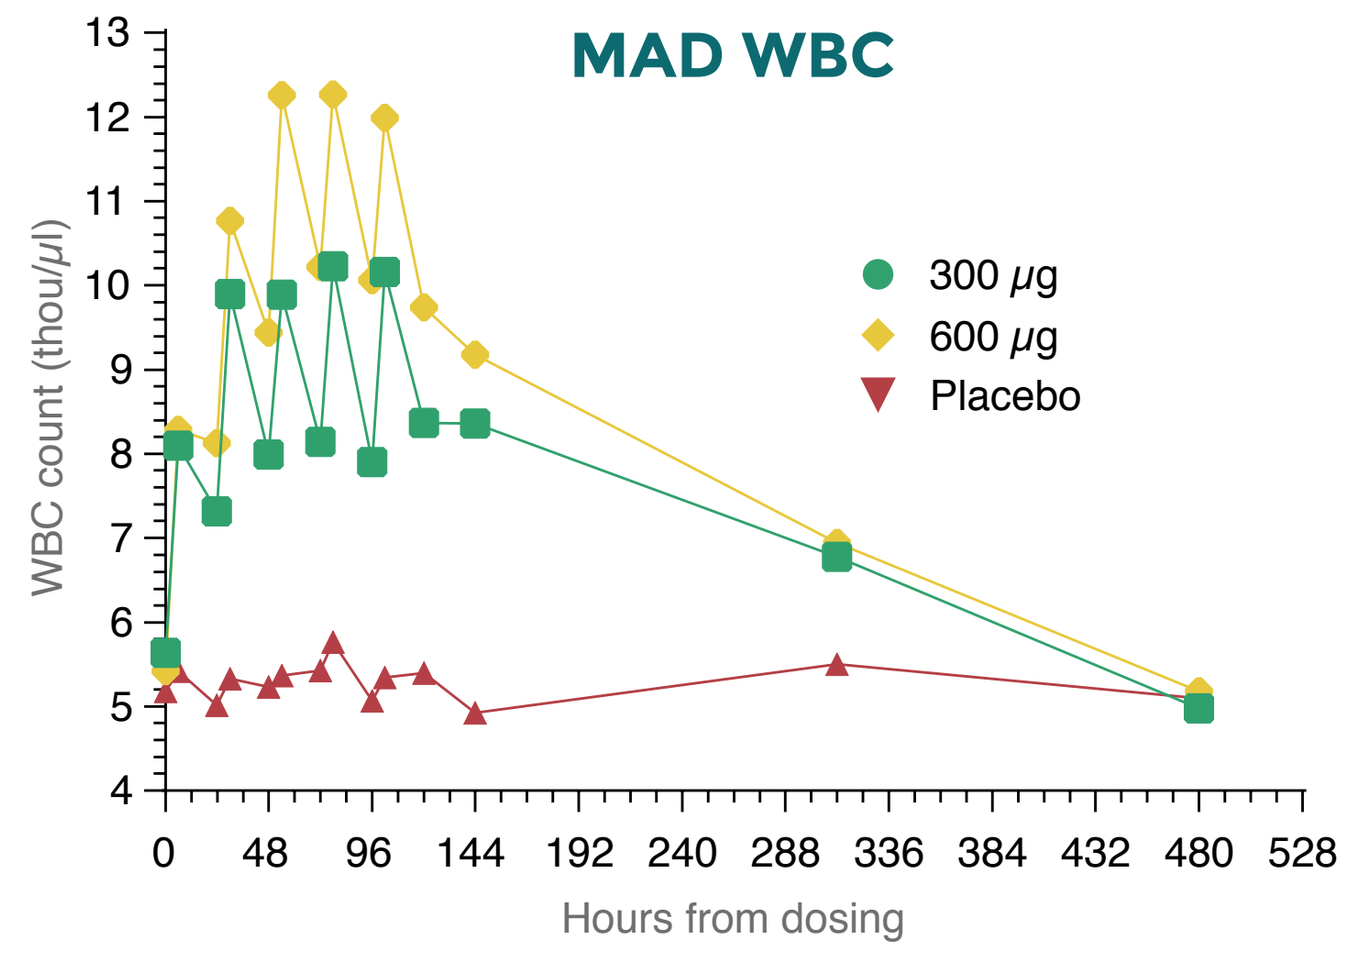
MAD WBC
WBC count (thou/µl)
Hours from dosing
300 µg
600 µg
Placebo

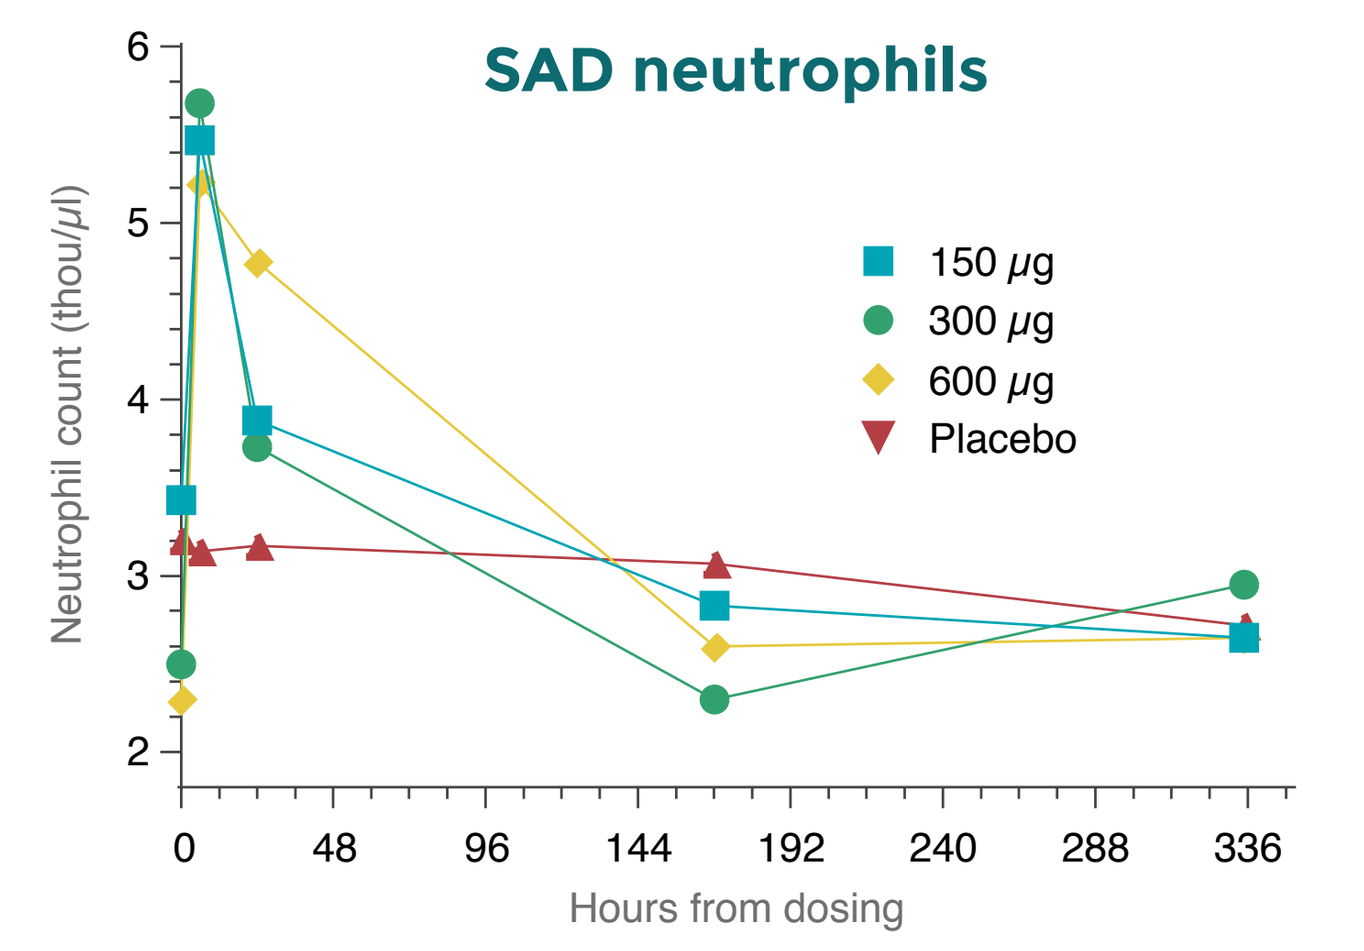
SAD neutrophils
Neutrophil count (thou/µl)
Hours from dosing
150 µg
300 µg
600 µg
Placebo

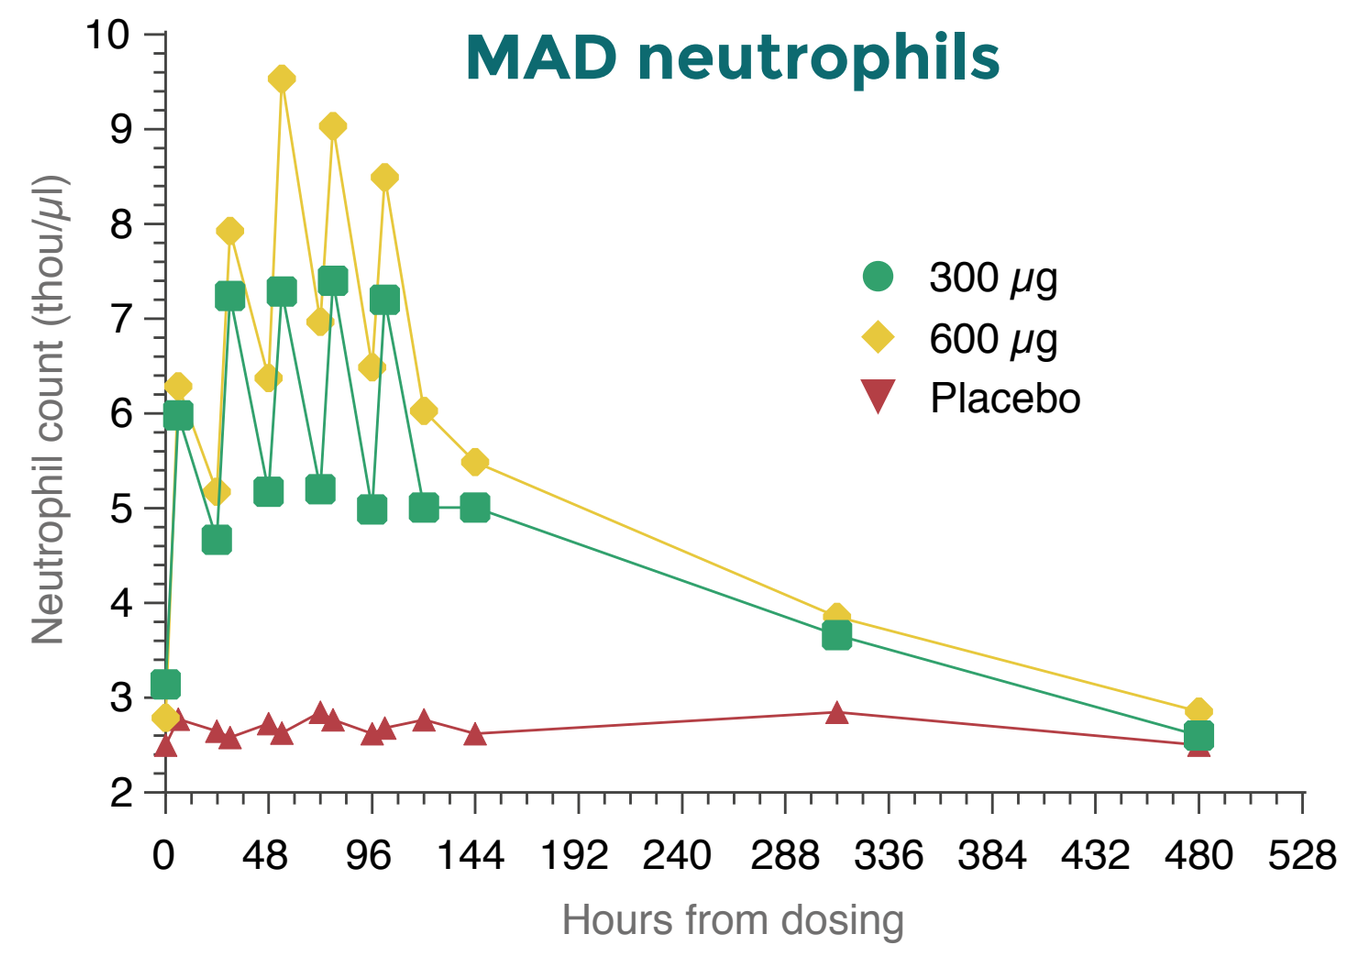
MAD neutrophils
Neutrophil count (thou/µl)
Hours from dosing
300 µg
600 µg
Placebo

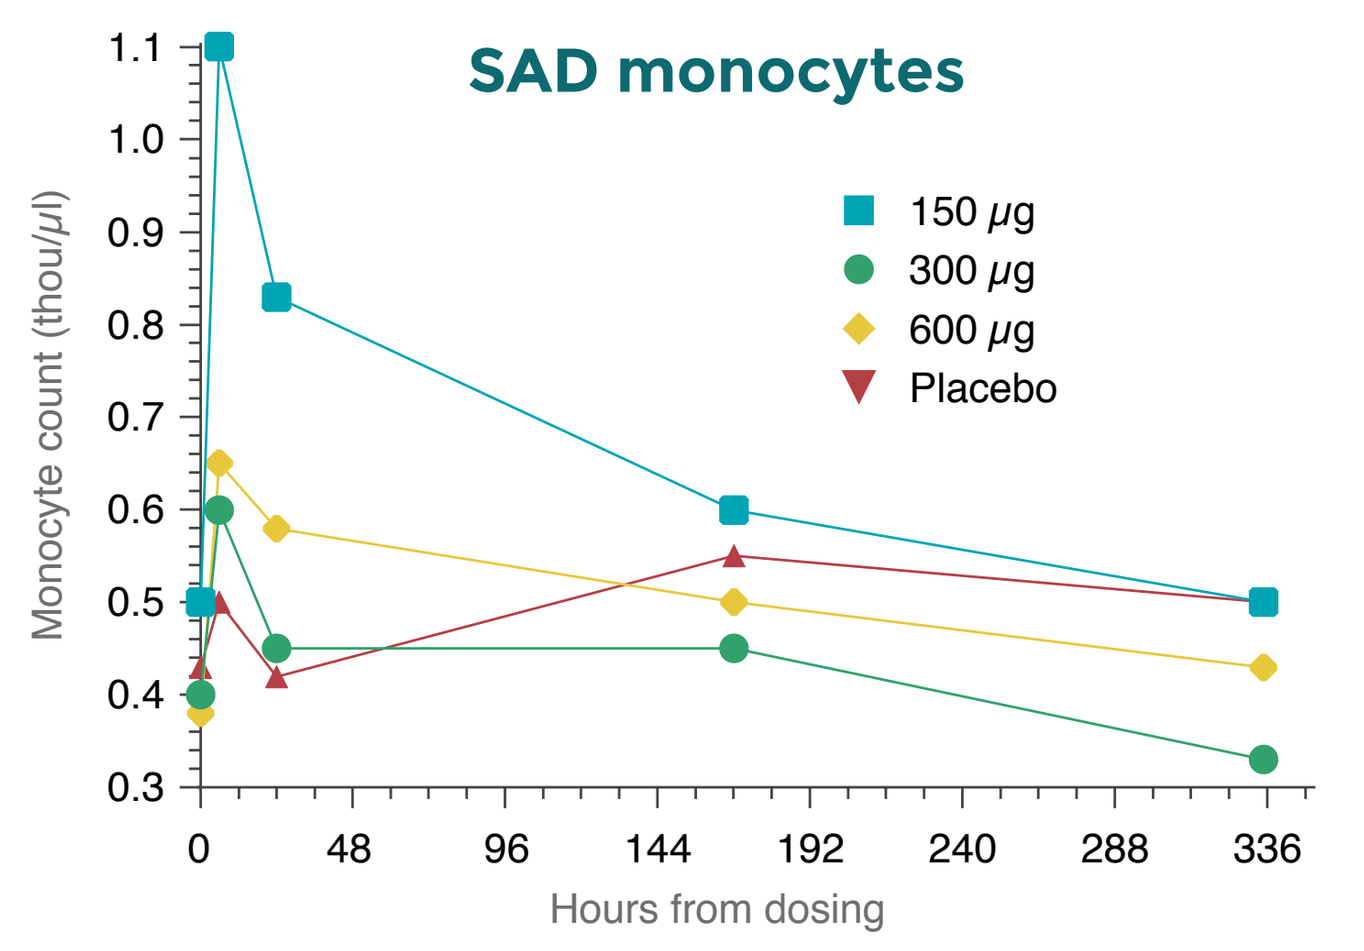
SAD monocytes
Monocyte count (thou/µl)
Hours from dosing
150 µg
300 µg
600 µg
Placebo

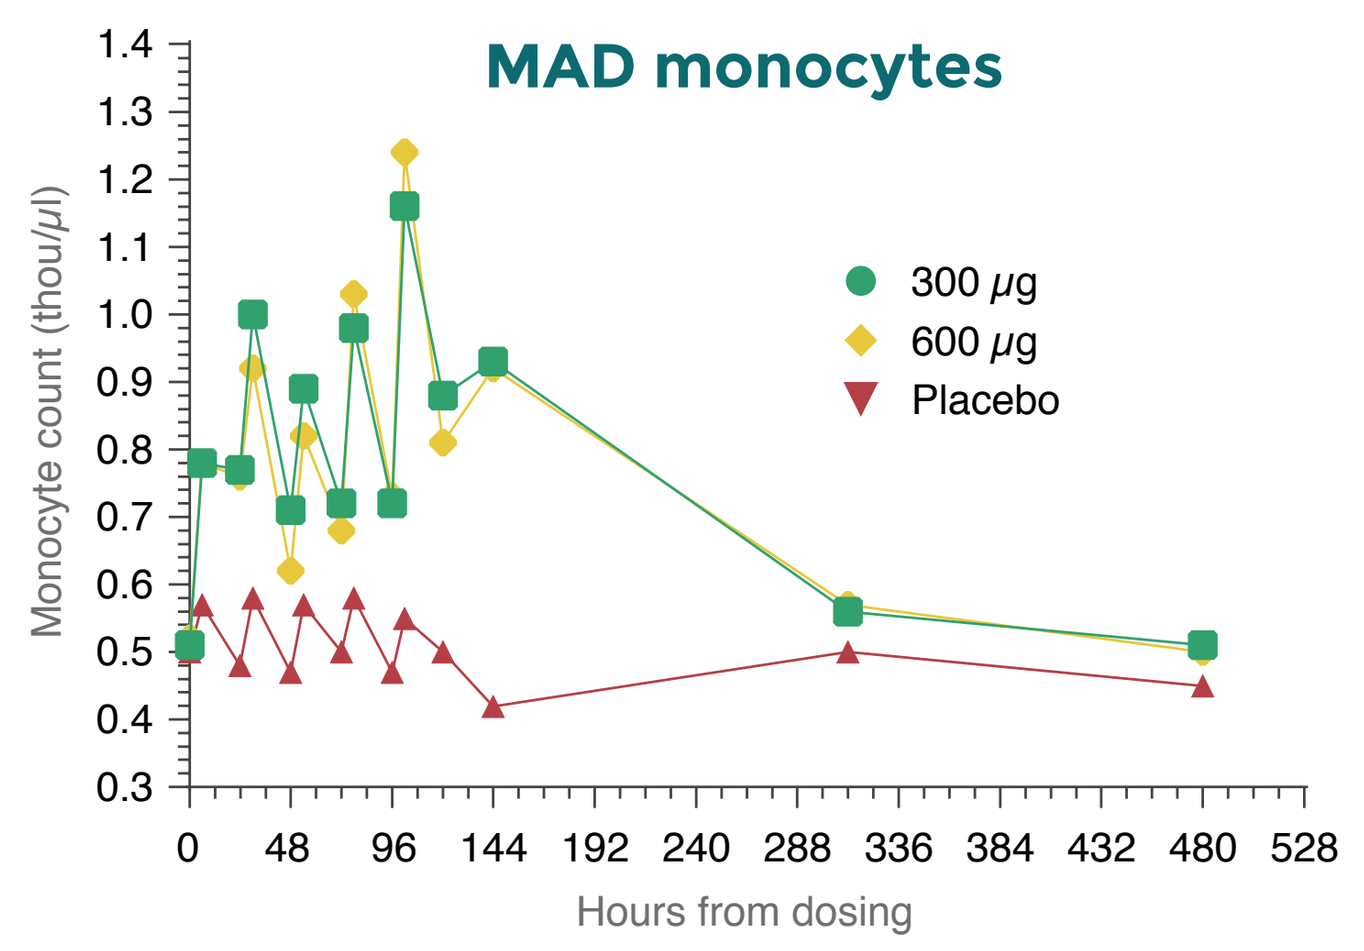
MAD monocytes
Monocyte count (thou/µl)
Hours from dosing
300 µg
600 µg
Placebo

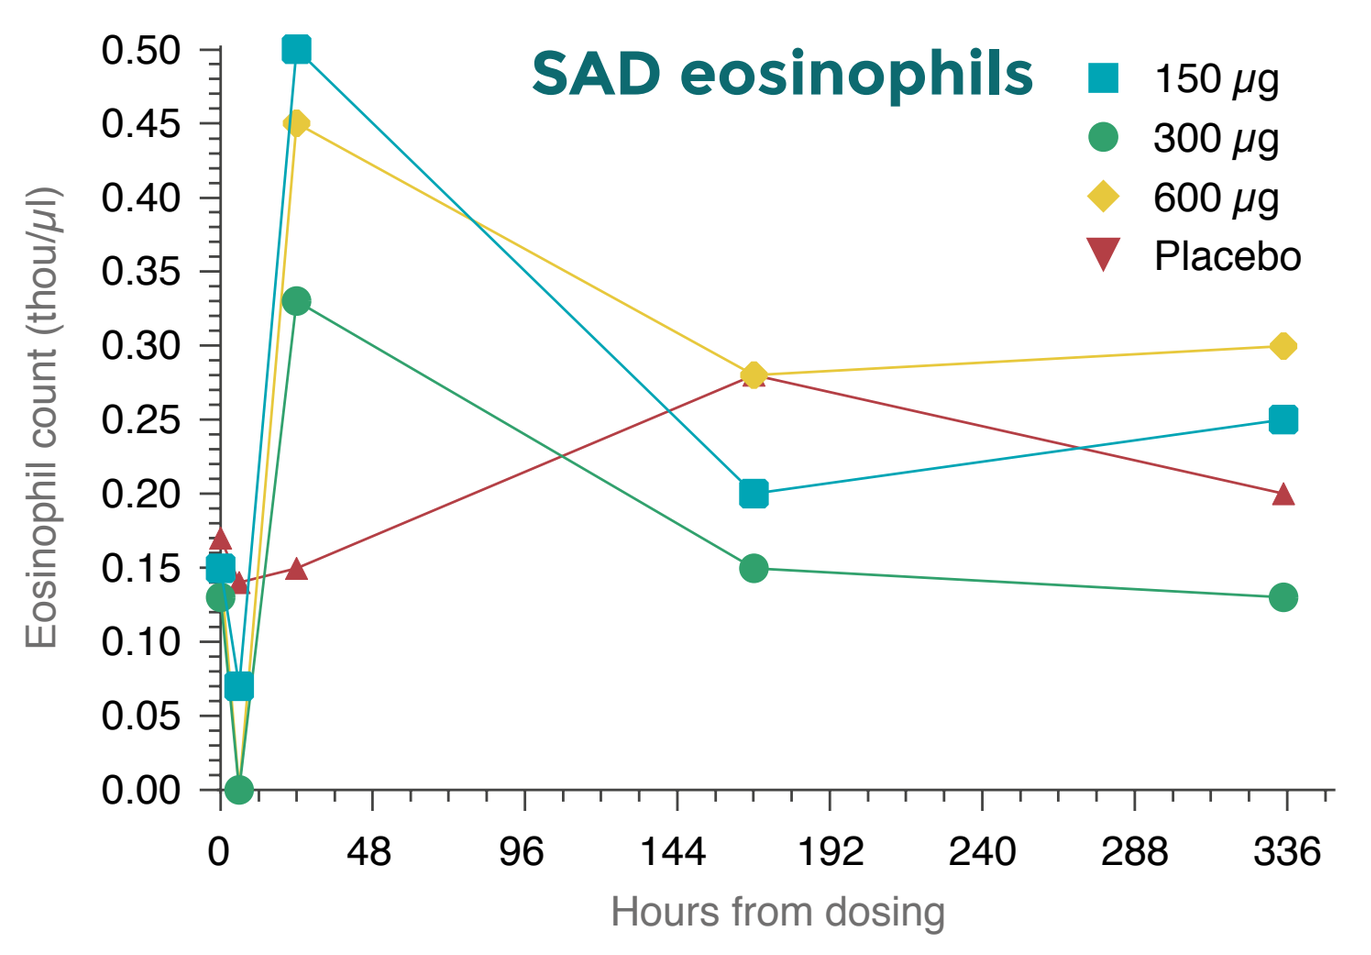
SAD eosinophils
Eosinophil count (thou/µl)
Hours from dosing
150 µg
300 µg
600 µg
Placebo

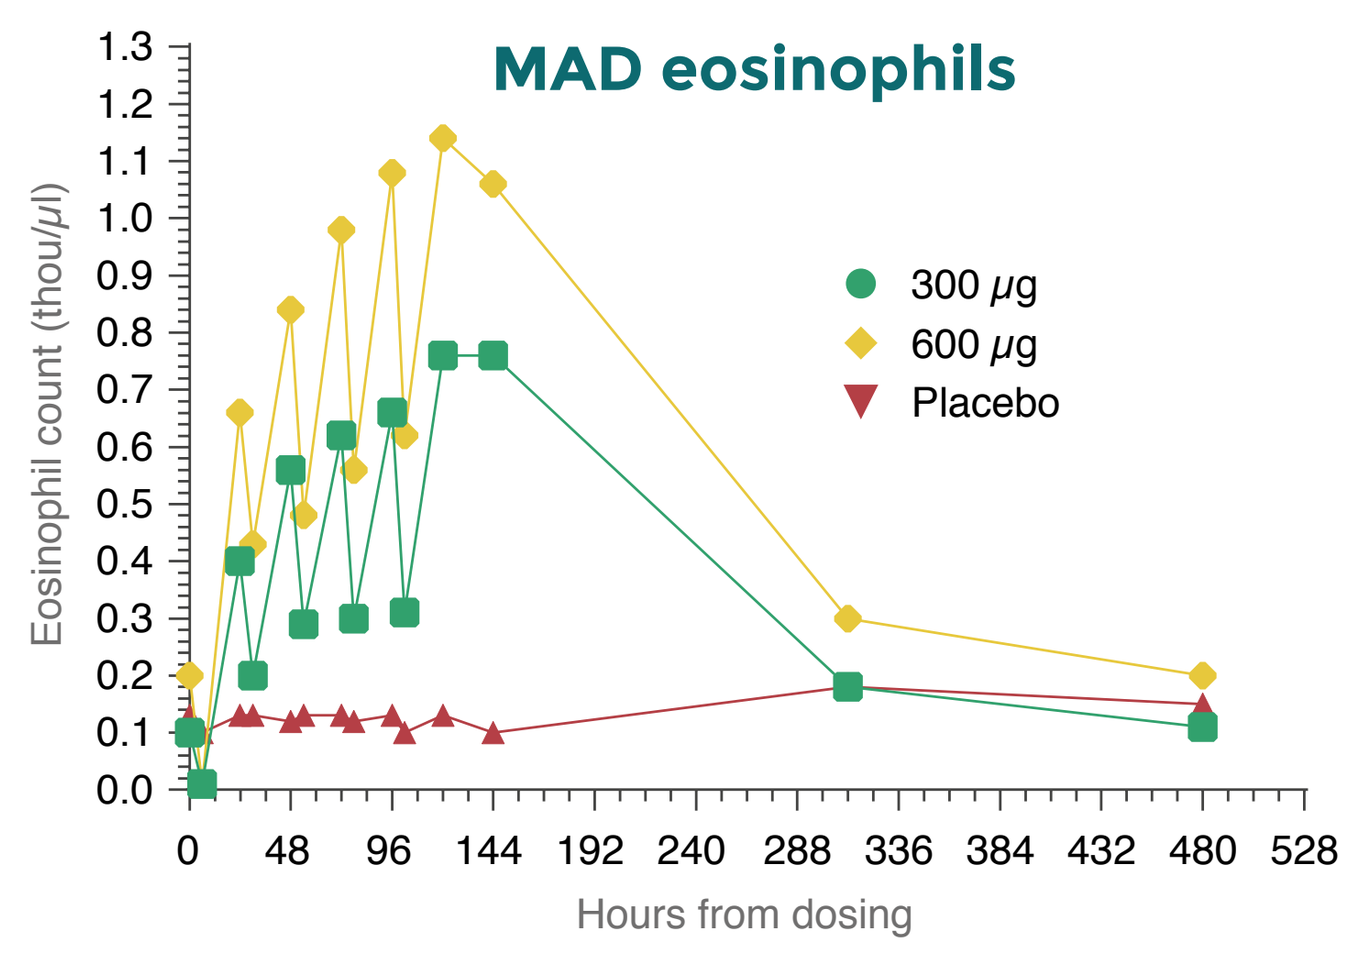
MAD eosinophils
Eosinophil count (thou/µl)
Hours from dosing
300 µg
600 µg
Placebo